RNAi Efficacy in Large and Small Fiber-Predominant Polyneuropathy of Hereditary Transthyretin Amyloidosis: HELIOS-A Post-Hoc Analysis

Violaine Planté-Bordeneuve[1], Andoni Echaniz-Laguna[2], Alejandra Gonzalez-Duarte[3,4], Karla Cárdenas-Soto[4], Hua Zheng[5], Amy Chan-Daniels[5], K. H. Vincent Lau[5], William Litchy[6]

[1]University Hospital Henri Mondor, Créteil, France; [2]Bicêtre University Hospital, Assistance Publique-Hôpitaux de Paris, Université Paris-Saclay, Le Kremlin-Bicêtre, France; [3]New York University Grossman School of Medicine, NYU Langone Health, New York, NY, USA; [4]Instituto Nacional de Ciencias Médicas y Nutrición Salvador Zubirán, Mexico City, Mexico; [5]Alnylam Pharmaceuticals, Cambridge, MA, USA; [6]Mayo Clinic, Rochester, MN, USA.

Scan to View
Country restrictions apply

Conclusions

- RNAi therapeutics vutrisiran and patisiran improved or prevented worsening of mNIS+7 regardless of baseline NCS sum scores
- In patients with early disease and minimal NCS abnormalities (NCS <2, less large-fiber involvement), patients had greater improvement in mNIS+7, primarily driven by QST. This finding in patients with less large-fiber involvement suggests predominant effects on small fibers
- In later disease (NCS ≥2), RNAi therapeutics address both fiber types, evidenced by effects on mNIS+7 and greater reductions in NfL, a marker for large-fiber neuropathy, compared with the NCS <2 group
- More patients in the baseline NCS <2 group showed potential disease reversal, supporting initiation of RNAi therapeutics as early as possible in the hATTR-PN disease course, reinforcing the importance of early diagnosis and treatment

Key takeaway

RNAi therapeutics vutrisiran and patisiran are effective for patients with hATTR-PN regardless of the degree of baseline nerve conduction study abnormalities

Introduction

hATTR-PN

- Hereditary transthyretin amyloidosis (hATTR) is a rapidly progressive, debilitating, fatal disease caused by variants of the *TTR* gene[1–3]
- Accumulation of misfolded TTR as amyloid deposits in the nerves, cardiac tissue, and other systems can manifest as polyneuropathy, cardiomyopathy, or a mixed phenotype[1–3]
- Polyneuropathy in patients with hATTR (hATTR-PN) is associated with various sensory, motor and autonomic nerve alterations.[3] It can involve both small and large nerve fibers, with small fibers often affected first[2,4,5]
- Large-fiber neuropathy may be diagnosed via clinical history, examination (e.g., motor weakness, abnormal vibratory sensations, and proprioception impairment) and/or nerve conduction study (NCS)[2,4,5]

Vutrisiran and Patisiran

- The RNAi therapeutics vutrisiran and patisiran decrease hepatic TTR synthesis and are approved for the treatment of hATTR-PN based on data from the phase 3 HELIOS-A (NCT03759379) and APOLLO (NCT01960348) studies, respectively[6–9]
- Vutrisiran and patisiran demonstrated robust treatment effects in patients with hATTR-PN across a broad range of disease severities, including those with mild disease[10,11]
- The outcomes of patients with varying degrees of NCS abnormalities at baseline, which may reflect disease stage and degree of large-fiber involvement, have not been examined independently

Objective

- This post-hoc analysis of HELIOS-A evaluated the efficacy of RNAi therapeutics in patients with hATTR-PN according to baseline NCS sum score (<2 vs ≥2) reflecting less vs more large-fiber involvement, respectively

Methods

- HELIOS-A was an international, multicenter, open-label, randomized, phase 3 study in patients with hATTR-PN in which patients were randomized 3:1 to receive vutrisiran (25 mg SC Q3M) or patisiran (0.3 mg/kg IV Q3W) for 18 months[11]
- In this post-hoc analysis, treatment effects at Month 18 were compared between patients with a baseline NCS sum score <2 vs ≥2
- NCS sum score <2 and NCS sum score ≥2 were calculated based on the sural SNAP, ulnar SNAP, ulnar CMAP, tibial CMAP, and peroneal CMAP amplitudes (range 0–2 for each component [0=normal, 1=mildly reduced, 2=very reduced], maximum score of 10)[12]

Results

Table 1. Demographics and Baseline Characteristics by Baseline NCS Sum Score Subgroup

	NCS Sum Score <2 (n=37)	NCS Sum Score ≥2 (n=127)
Age, years, median (range)	60.0 (37.0–80.0)	60.0 (26.0–85.0)
Male, n (%)	18 (48.6)	88 (69.3)
Race, n (%)		
Asian	4 (10.8)	25 (19.7)
Black or African American	1 (2.7)	7 (5.5)
White	26 (70.3)	89 (70.1)
Multiple	1 (2.7)	0
Other	5 (13.5)	6 (4.7)
Time since hATTR diagnosis, years, median (range)	2.4 (0.1–13.4)	2.2 (0.0–15.3)
Genotype, n (%)		
V30M	13 (35.1)	61 (48.0)
Non-V30M	24 (64.9)	66 (52.0)
BMI, kg/m^2, mean (SD)	27.0 (4.4)	23.8 (4.9)
mNIS+7 score, mean (SD)	27.4 (22.9)	69.3 (32.7)
Norfolk QoL-DN score, mean (SD)	33.1 (28.4)[a]	51.2 (25.6)
10-MWT, mean (SD)	1.236 (0.298)	0.940 (0.394)
NfL (pg/mL), mean (SD)	25.9 (30.0)	69.1 (39.9)[b]

[a]n=36; [b]n=110.

Results

- At M18 in the overall population, mNIS+7 improved in NCS <2 patients (n=36) vs baseline (mean [SD] change –5.7 [13.1], *P*=0.013) and was maintained in NCS ≥2 patients (n=112; mean [SD] change 2.6 [16.4], *P*=0.102)
 - For reference, in the placebo group of the APOLLO study (n=77), least-squares mean (SE) change from baseline in mNIS+7 at Month 18 was 28 (2.6)[10]
- The efficacy of RNAi therapeutics was generally consistent between patients with high and low baseline NCS across measures of polyneuropathy

Figure 1. Proportion of Patients With Potential Disease Reversal, Defined as Decrease From Baseline at Month 18 in mNIS+7 Score or in the Norfolk QoL-DN Score, According to Baseline NCS Sum Score Subgroup

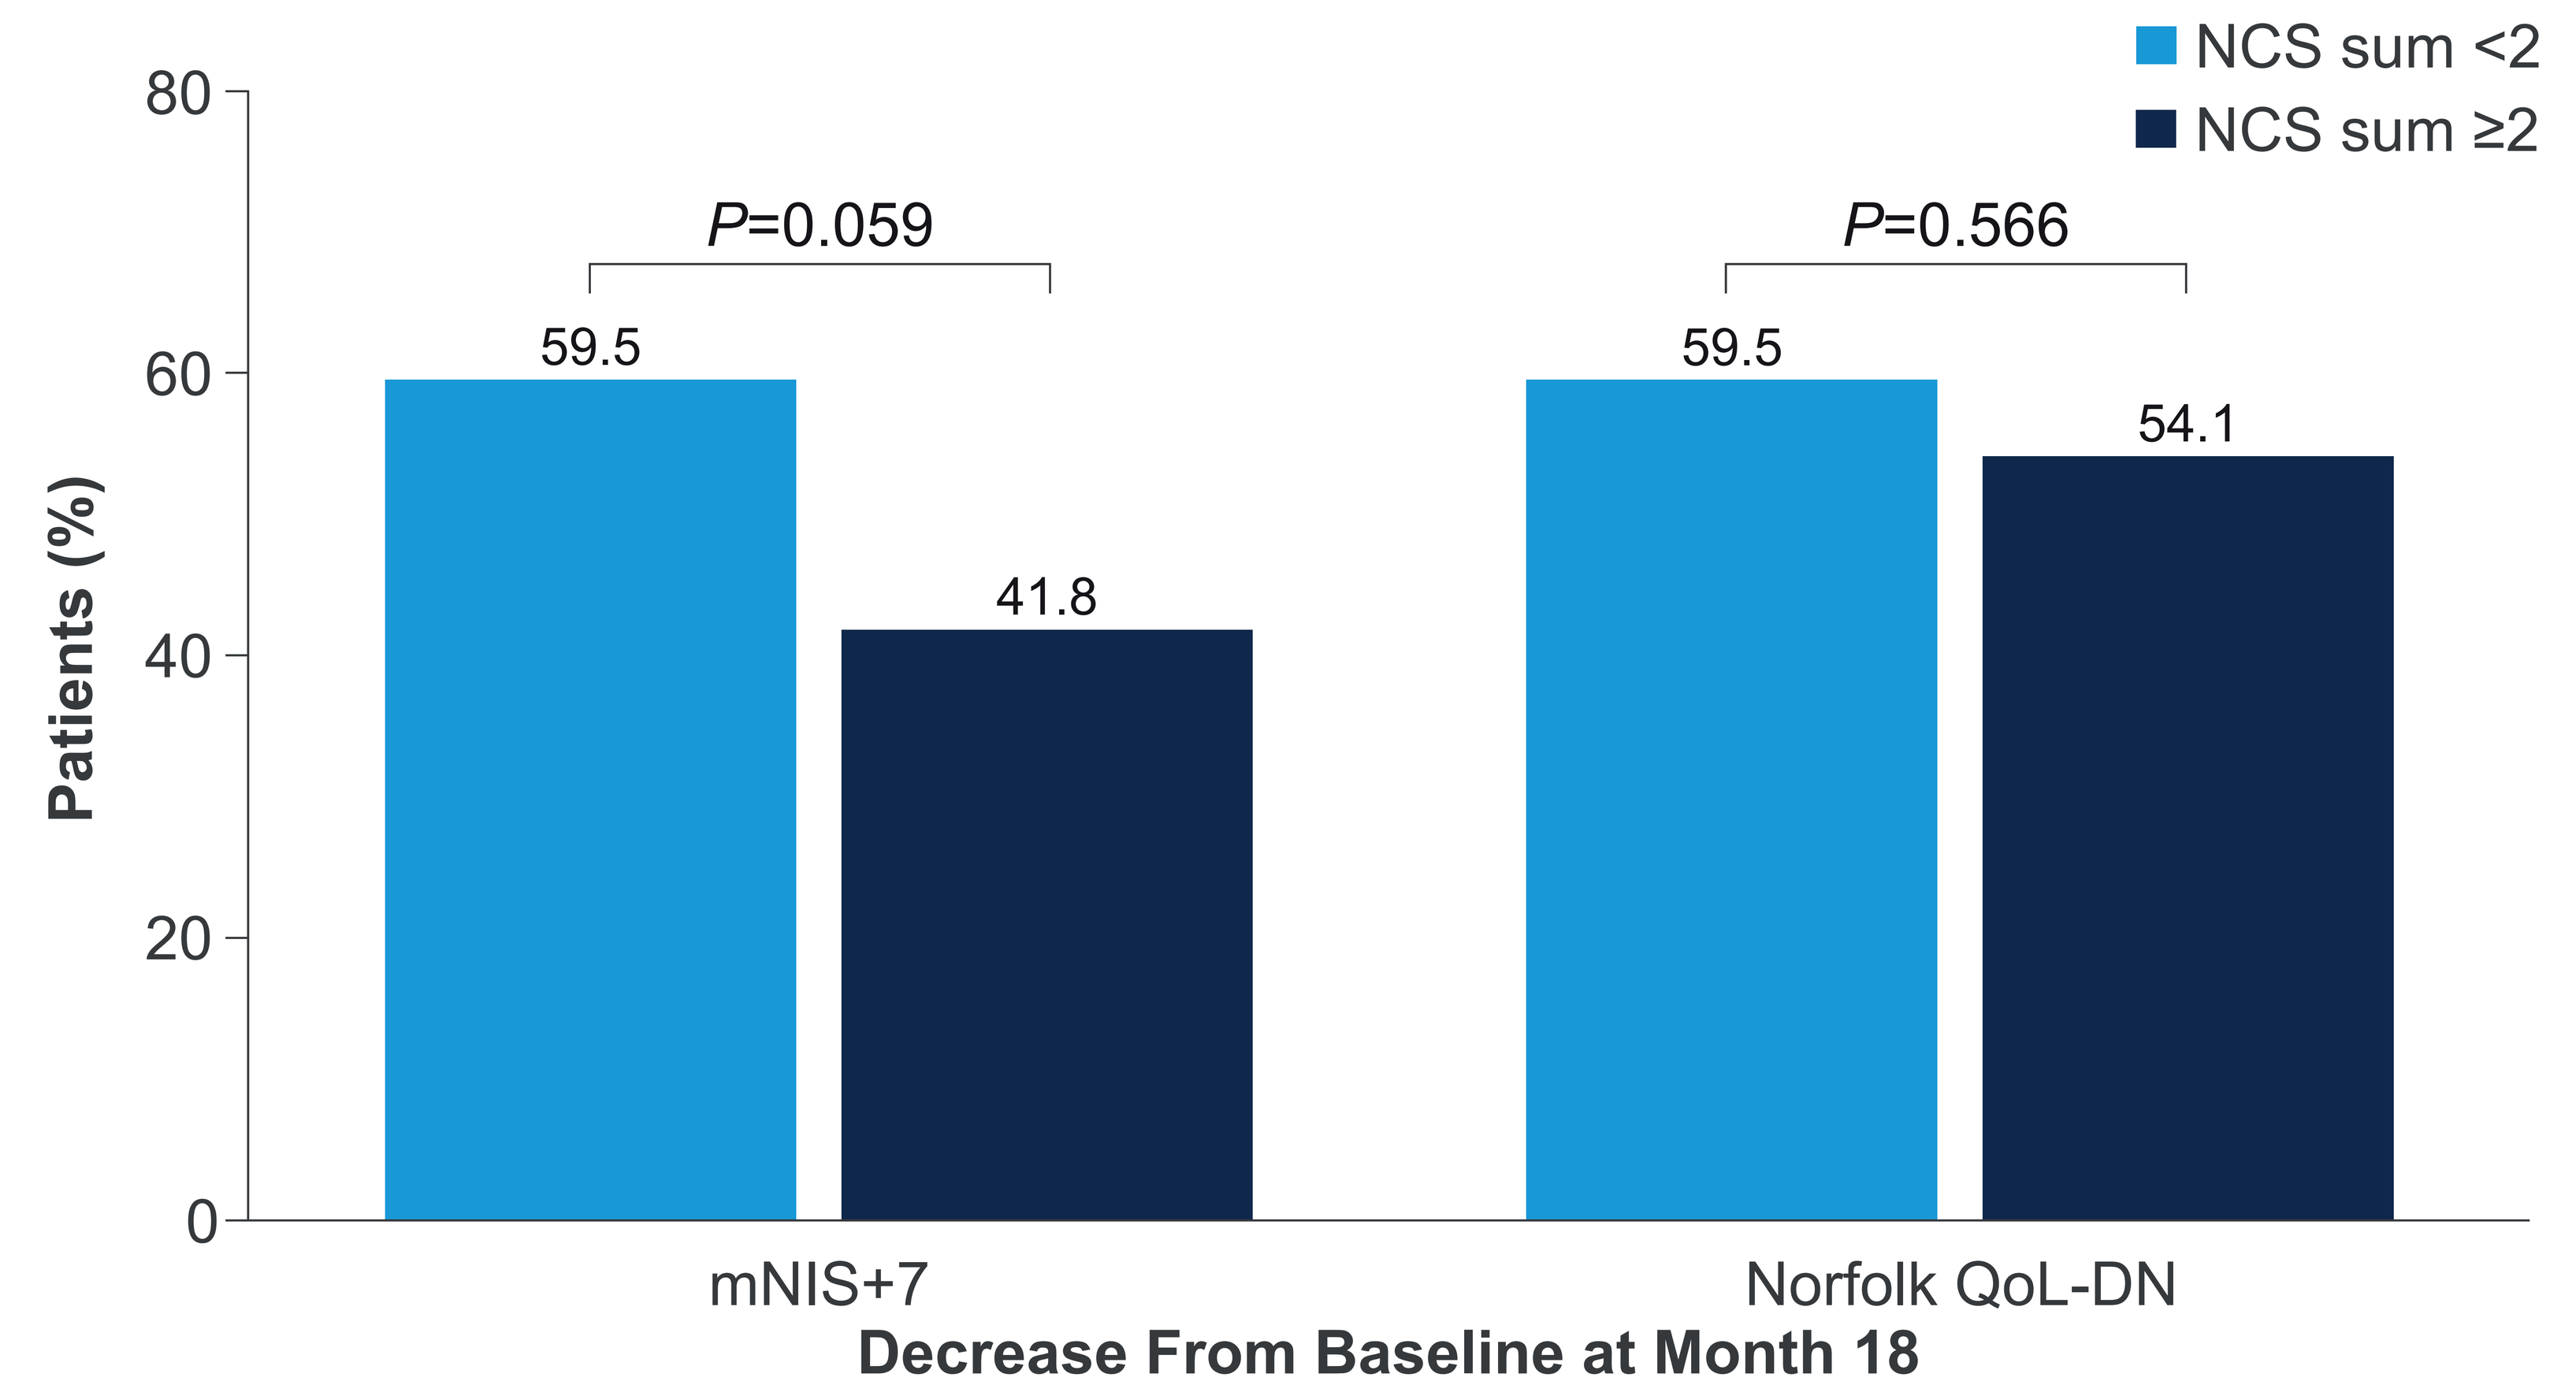
NCS sum <2
NCS sum ≥2
80
60
40
20
0
Patients (%)
P=0.059
P=0.566
59.5
41.8
59.5
54.1
mNIS+7
Norfolk QoL-DN
Decrease From Baseline at Month 18

Figure 2. Forest Plot of Change From Baseline in mNIS+7 Individual Components At Month 18 by Baseline NCS Sum Score Subgroup

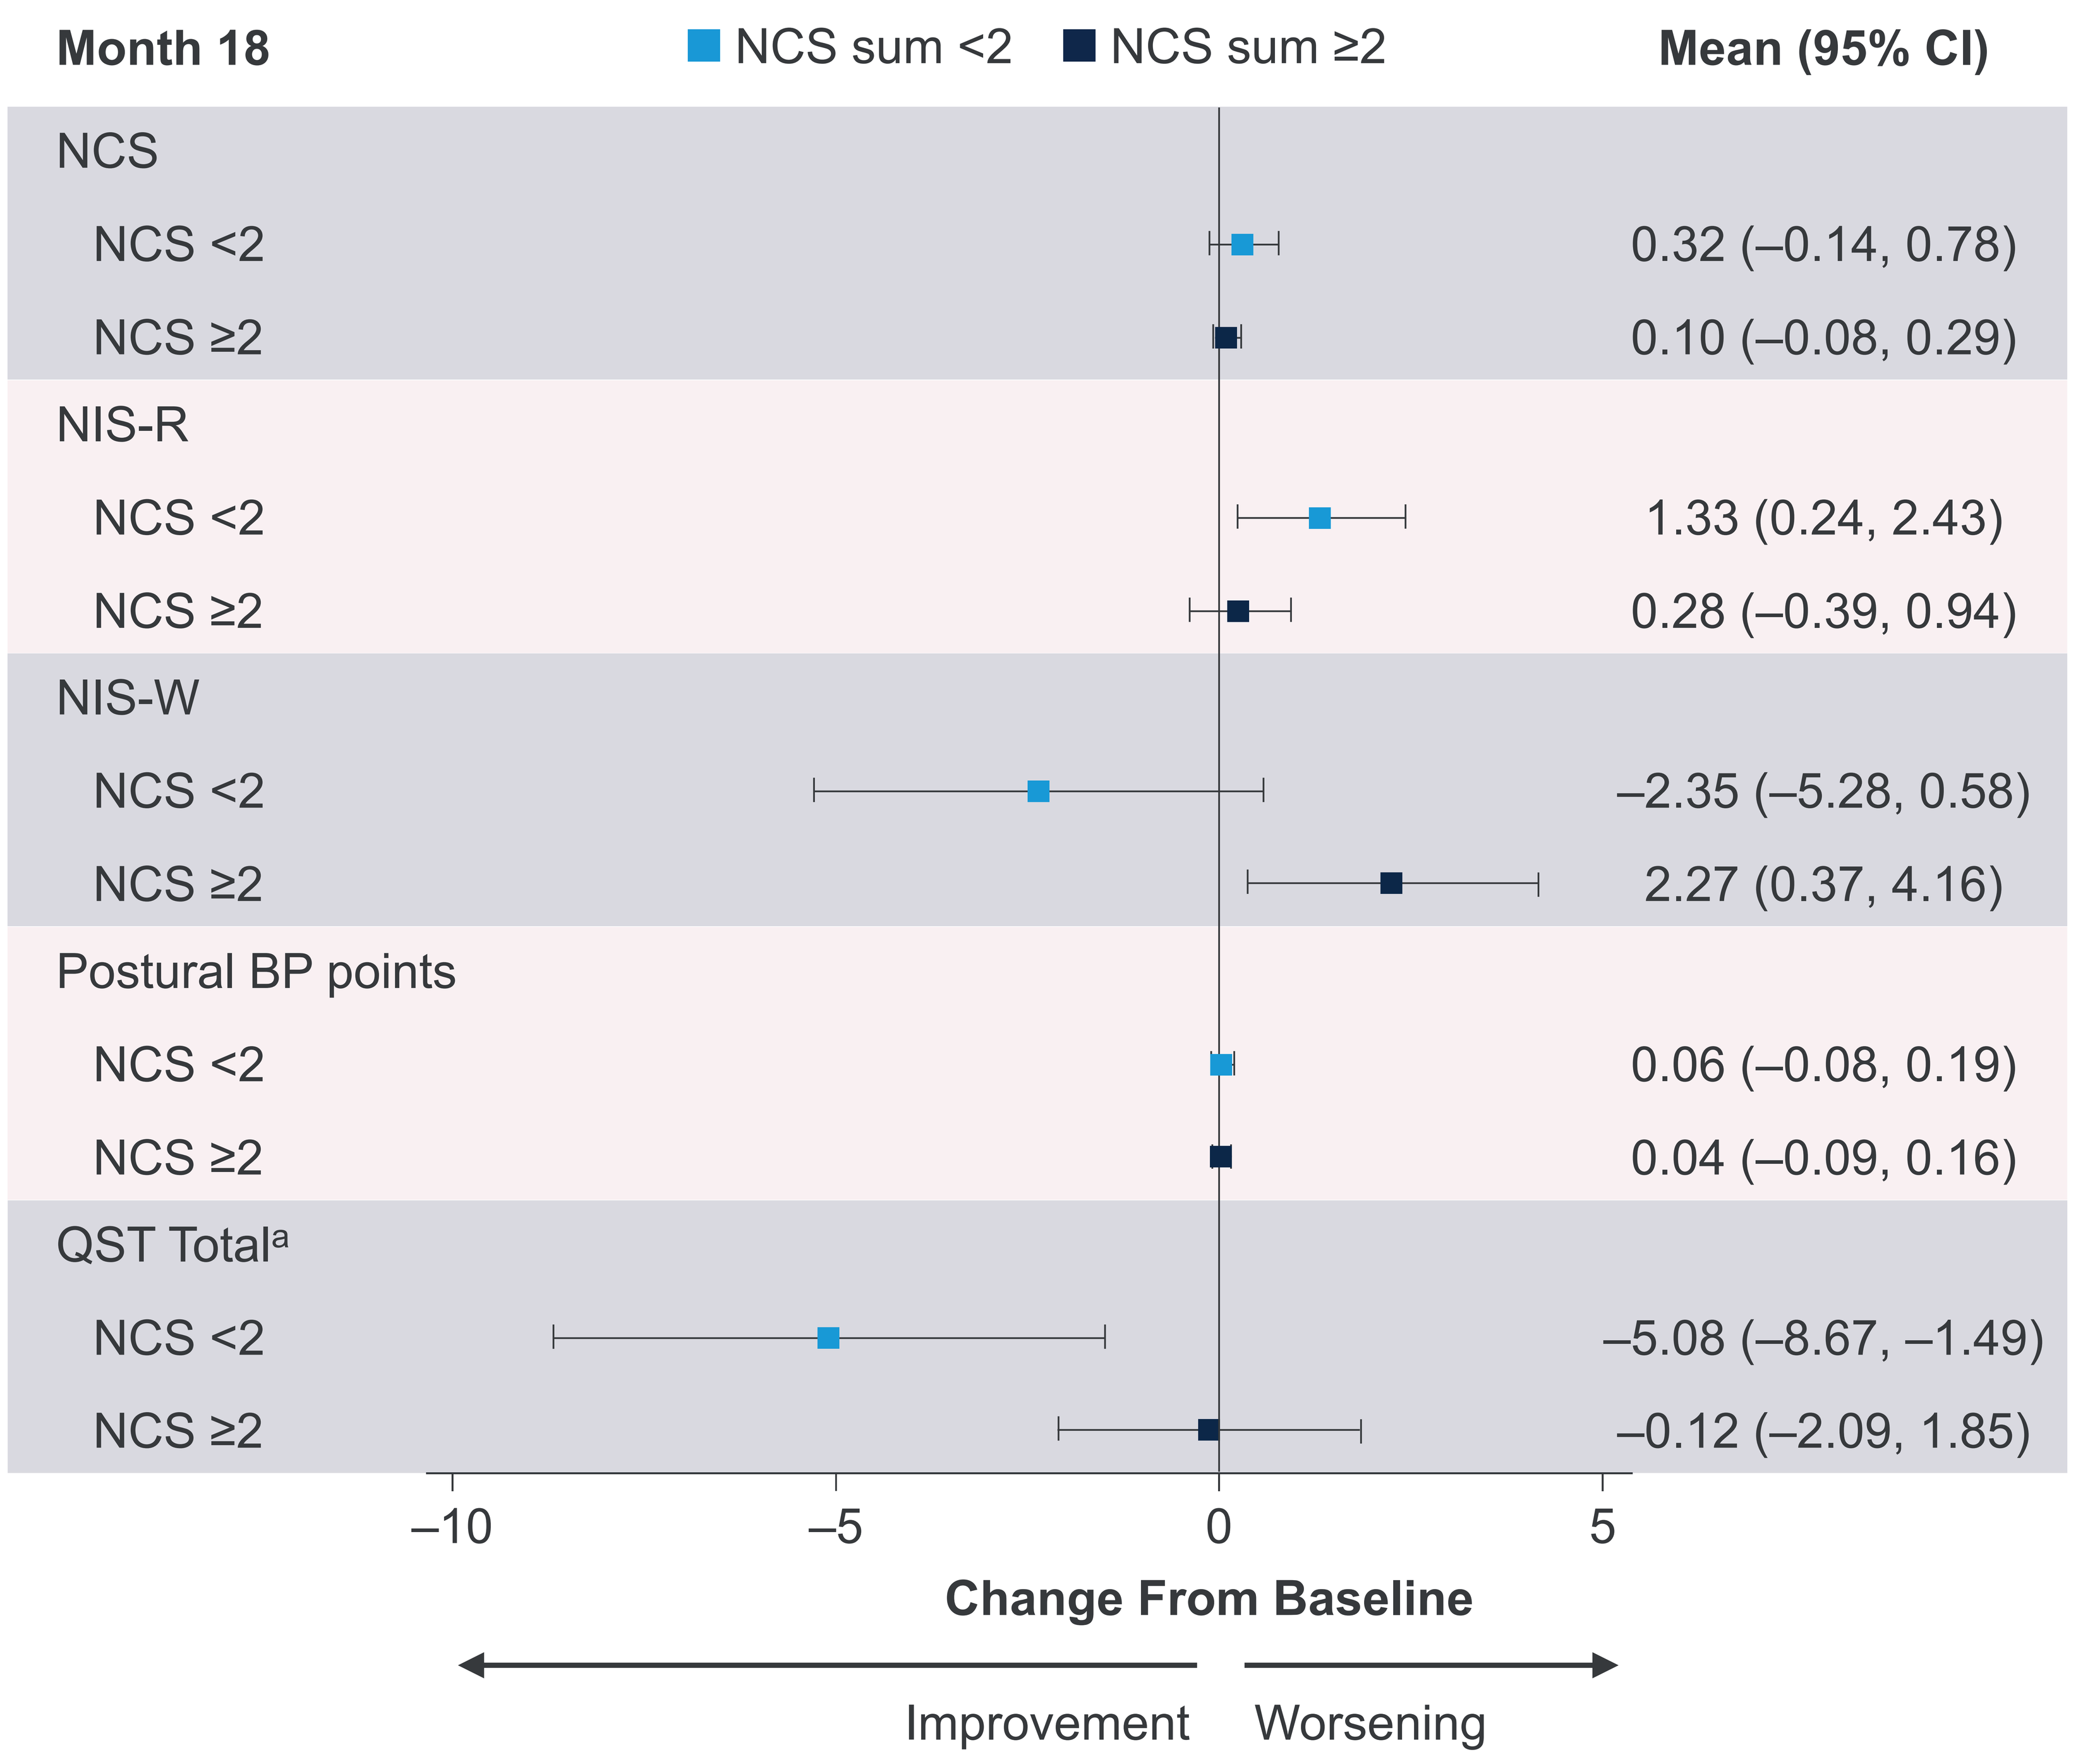
Month 18
NCS sum <2
NCS sum ≥2
Mean (95% CI)
NCS
NCS <2
0.32 (–0.14, 0.78)
NCS ≥2
0.10 (–0.08, 0.29)
NIS-R
NCS <2
1.33 (0.24, 2.43)
NCS ≥2
0.28 (–0.39, 0.94)
NIS-W
NCS <2
–2.35 (–5.28, 0.58)
NCS ≥2
2.27 (0.37, 4.16)
Postural BP points
NCS <2
0.06 (–0.08, 0.19)
NCS ≥2
0.04 (–0.09, 0.16)
QST Total[a]
NCS <2
–5.08 (–8.67, –1.49)
NCS ≥2
–0.12 (–2.09, 1.85)
–10
–5
0
5
Change From Baseline
Improvement
Worsening

[a]QST measures heat pain and touch pressure from 10 distributed anatomic sites on one side of the body. Scoring is based on point abnormality (<95th=0 point, ≥95th–<99th=1 point, and ≥99th=2 points). The test score is 2 × the total scores for QST of heat pain by body surface area and QST of touch pressure by body surface area. The maximum total score for QST is 80.

Table 2. Change From Baseline in Norfolk QoL-DN Score, 10-MWT, and NfL Levels at Month 18 by Baseline NCS Sum Score Subgroups

Mean (SD)	NCS Sum Score <2 (n=37)	NCS Sum Score ≥2 (n=127)
Norfolk QoL-DN score		
n	35	114
Change from baseline	–4.0 (23.0)	–1.3 (19.6)
P value	0.501	
10-MWT speed (m/s)		
n	36	114
Change from baseline	0.026 (0.268)	–0.038 (0.261)
P value	0.204	
NfL (pg/mL)		
n	35	100
Change from baseline	–3.6 (15.3)	–24.6 (34.4)
P value	<0.001	

We thank the patients, their families, investigators, staff and collaborators for their participation in HELIOS-A.

Acknowledgments: Medical writing support, under the guidance of the authors, was provided by Elizabeth Drysdale, MBChB, and Corinne S Wilson, employees from the Publications and Medical Affairs Division of Omnicom Health Medical Communications, and was funded by Alnylam Pharmaceuticals in accordance with Good Publication Practice, GPP 2022 (*Ann Intern Med* 2022;175:1298–304).

Funding: This study was funded by Alnylam Pharmaceuticals.

If you are seeking additional scientific information related to Alnylam therapeutics, non-US HCPs should contact medinfo@alnylam.com. US HCPs may visit the Alnylam US Medical Affairs website at RNAiScience.com.

Disclosures: VPB reports consulting fees from Alnylam Pharmaceuticals, AstraZeneca, and Purpose Pharma; **AEL** reports consulting fees from Alnylam Pharmaceuticals, Argenx, BridgeBio, Intellia Therapeutics, Pfizer Inc, and Sanofi; **AGD** reports consulting fees from Alnylam Pharmaceuticals, AstraZeneca, BridgeBio, Intellia Therapeutics, and Pfizer Inc; **KCS** reports consulting fees from AstraZeneca and Intellia Therapeutics; **HZ, ACD,** and **VL** are employees of Alnylam Pharmaceuticals, and report ownership of equity in Alnylam Pharmaceuticals; **WL** reports consulting fees from Alexion Pharmaceuticals, Alnylam Pharmaceuticals, Intellia Therapeutics, and Ionis Pharmaceuticals.

Presented at: Peripheral Nerve Society (PNS) Annual Meeting, Jun 13–16, 2026, Maastricht, The Netherlands.

References: 1. Gonzalez-Duarte, Ulloa-Aguirre. *Int J Mol Sci* 2021;22:13158; 2. Adams et al. *Nat Rev Neurol* 2019;15:387–404; 3. Nativi-Nicolau et al. *Heart Fail Rev* 2022;27:785–93; 4. Tozza et al. *J Peripher Nerv Syst* 2021;26:155–59; 5. Waddington-Cruz et al. *J Peripher Nerv Syst* 2021;26:160–16; 6. Alnylam Pharmaceuticals. Onpattro. Prescribing Information 2025. Available from: https://www.alnylam.com/sites/default/files/pdfs/ONPATTRO-Prescribing-Information.pdf; 7. Alnylam Pharmaceuticals. Amvuttra. Prescribing information 2025 [accessed 2026 April 15]. Available from: https://www.alnylam.com/sites/default/files/pdfs/amvuttra-us-prescribing-information.pdf; 8. European Medicines Agency. Amvuttra. Summary of Product Characteristics 2025 [accessed 2026 April 15]. Available from: https://www.ema.europa.eu/en/documents/product-information/amvuttra-epar-product-information_en.pdf; 9. European Medicines Agency. Onpattro. Summary of Product Characteristics 2025 [accessed 2025 December 12]. Available from: https://www.ema.europa.eu/en/documents/product-information/onpattro-epar-product-information_en.pdf; 10. Adams et al. *N Engl J Med* 2018;379:11–21; 11. Adams et al. *Amyloid* 2023;30:18–26; 12. Dyck et al. *J Neurol Sci* 2019;405:116424.

Abbreviations: 10-MWT, 10-metre walk test; BMI, body mass index; BP, blood pressure; CI, confidence interval; CMAP, compound muscle action potential; hATTR, hereditary transthyretin amyloidosis; hATTR-PN, hereditary transthyretin amyloidosis with polyneuropathy; IV, intravenous; mNIS+7, modified neuropathy impairment score+7; NCS, nerve conduction study; NfL, neurofilament light chain; NIS, neuropathy impairment score; NIS-R, neuropathy impairment score-reflexes; NIS-W, neuropathy impairment score-weakness; QoL, quality of life; QoL-DN, Quality of Life-Diabetic Neuropathy questionnaire; QST, quantitative sensory testing; Q3M, every three months; Q3W, every three weeks; RNAi, RNA interference; SC, subcutaneous; SD, standard deviation; SNAP, sensory nerve action potential; TTR, transthyretin.